316

Autoantibody Signatures in Chronic Inflammatory Demyelinating Polyradiculoneuropathy: Insights on Glycolipid Reactivity From the ADHERE Trial

Susan K. Halstead,[1] Hugh J. Willison,[1] Anneleen Remmerie,[2] Erik Hofman,[2] Geoffrey Istas,[2] Bianca Balbino[2]

[1]School of Infection and Immunity, University of Glasgow, Glasgow, UK; [2]argenx, Ghent, Belgium

BACKGROUND

- CIDP is a rare, immune-mediated neuropathy characterized by progressive muscle weakness and sensory dysfunction[1,2]
- Evidence supports a role for pathogenic IgG in CIDP, although pathogenic autoantibodies have not been identified in most patients[3–6]
- Approximately 40% of patients have antibodies against myelinated nerve components, although the nature of the antigens is unknown[7–9]
- Glycolipids enriched in myelinated nerves include galactocerebroside and sulfatide[10]
- Anti-glycolipid antibodies are uncommonly detected in patients with CIDP, but have been identified as targets in other neurological diseases such as Guillain-Barré syndrome[11]
- Efgartigimod, a human IgG1 Fc fragment and a natural ligand of the FcRn, prevents IgG recycling and increases its lysosomal degradation, without impacting IgG function or production[12]
- In the ADHERE trial (NCT04281472), efgartigimod SC (co-formulated with recombinant human hyaluronidase PH20) reduced relapse risk and was well tolerated in participants with CIDP[13]

FIGURE 1 Glycoarray Antigen Format

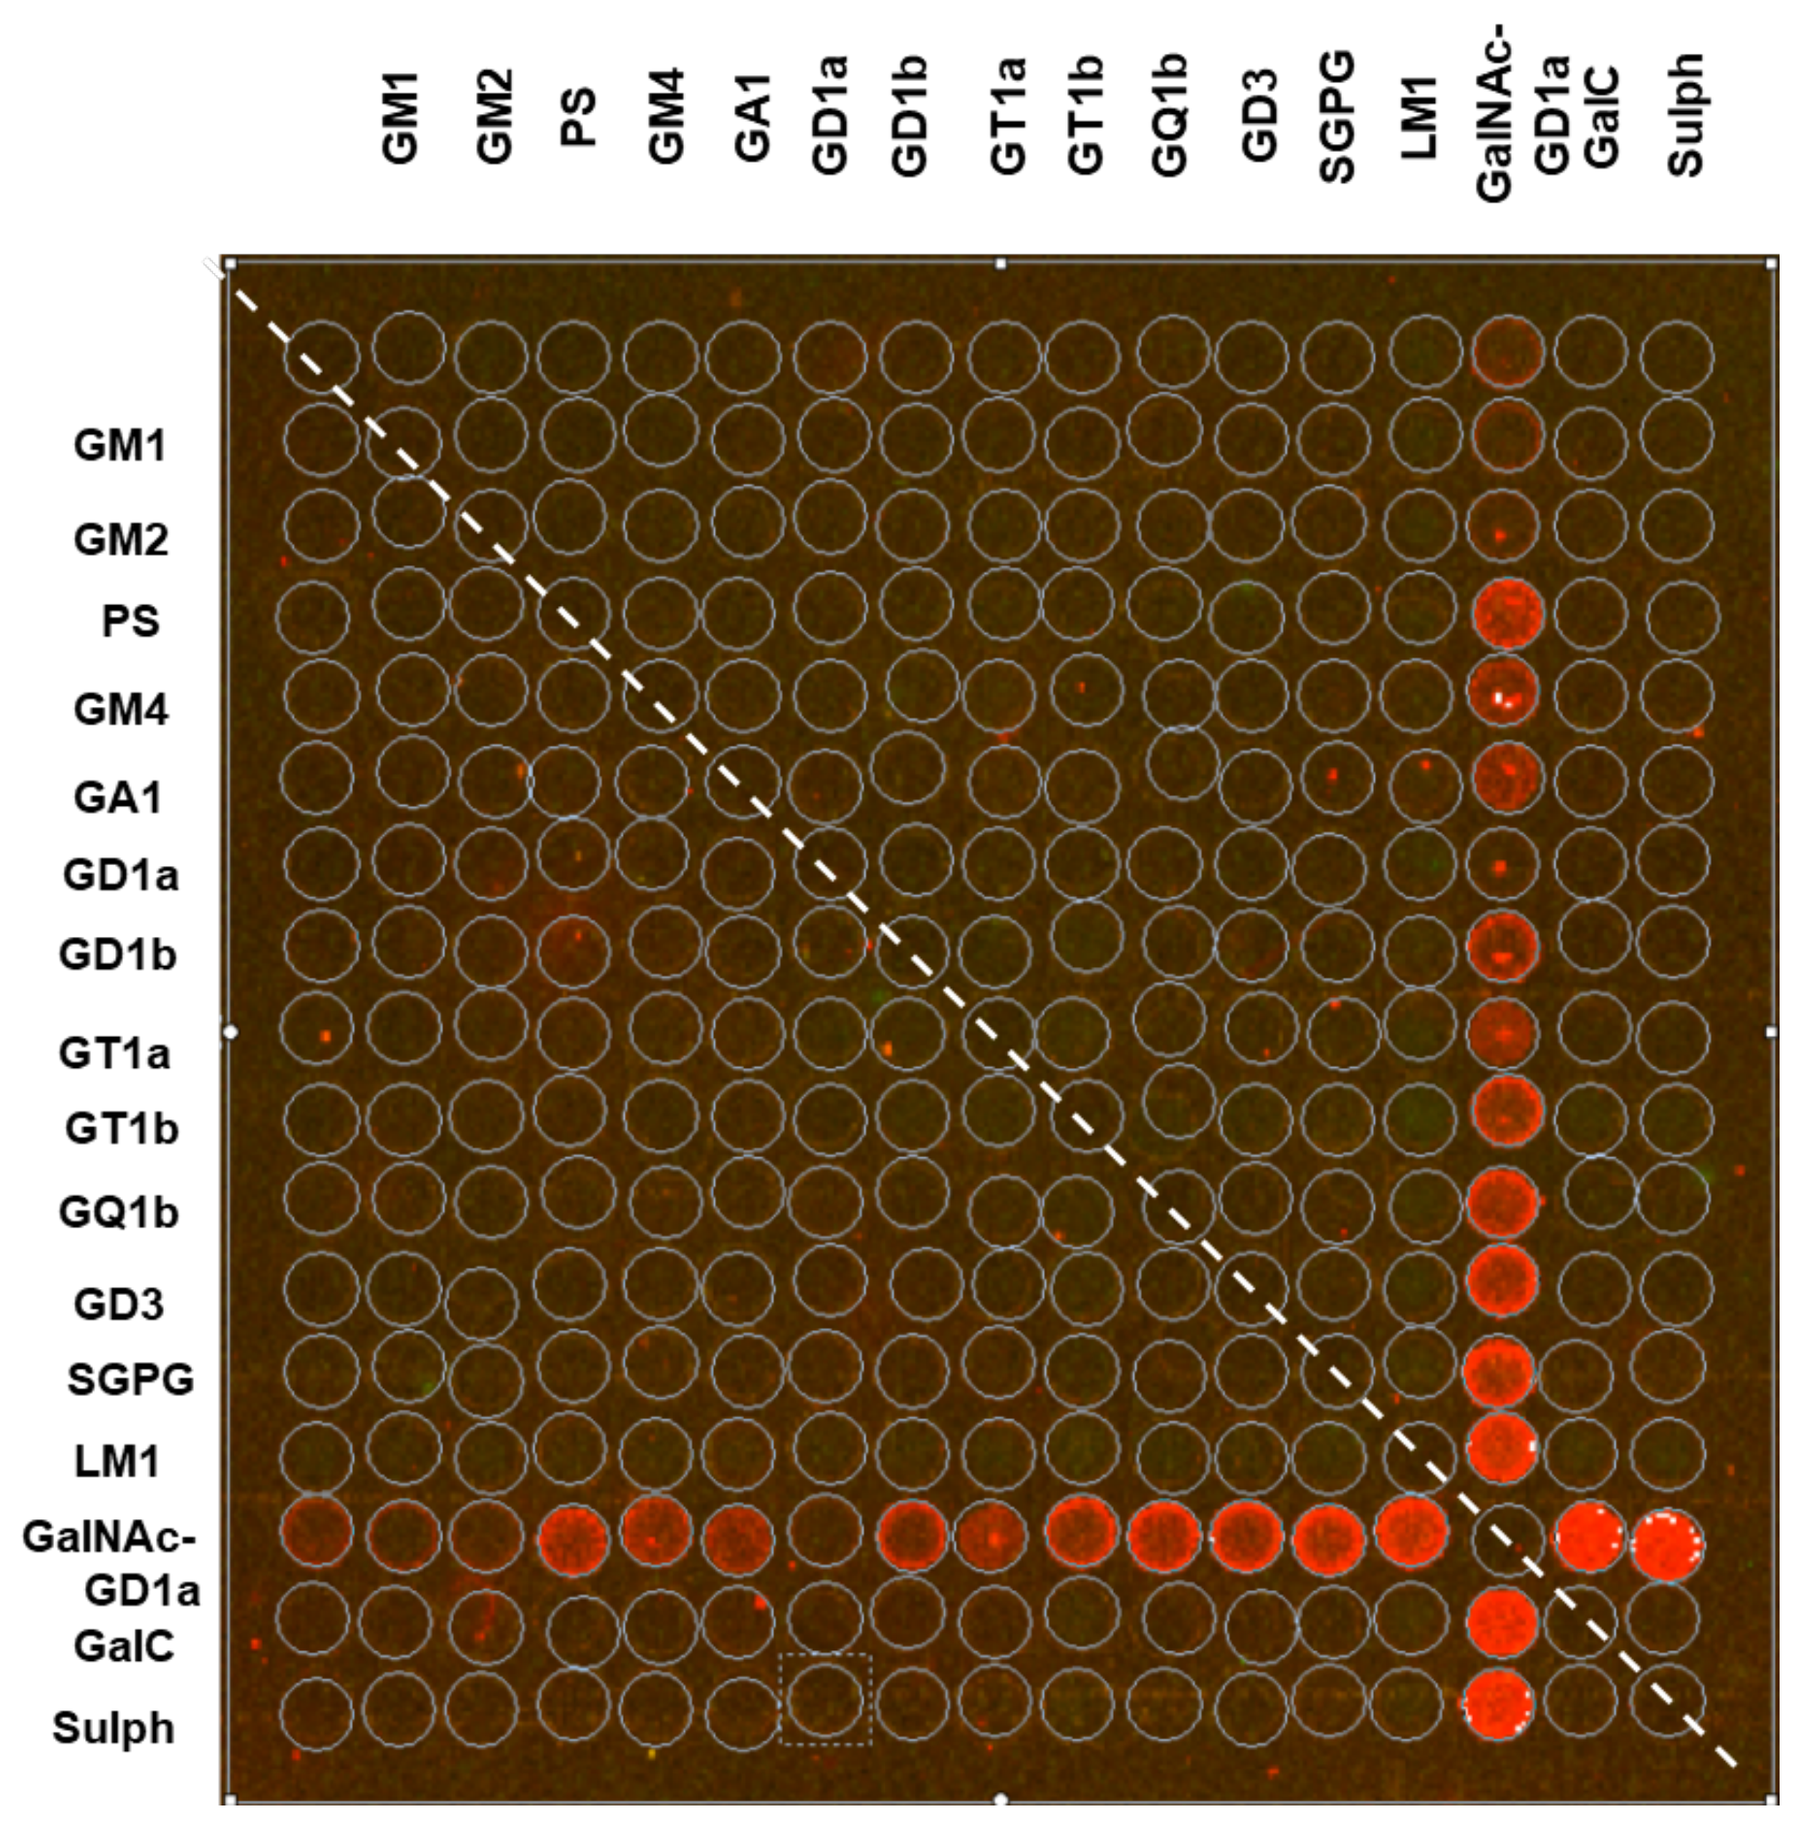
GM1
GM2
PS
GM4
GA1
GD1a
GD1b
GT1a
GT1b
GQ1b
GD3
SGPG
LM1
GalNAc-GD1a
GalC
Sulph

Example of glycoarray antigen format. Antigen targets are printed in duplicate with a line of symmetry (solvent only) running diagonally from top left to bottom right. The first row and column of the grid-format contain single antigens only, with all remaining coordinates containing 1:1 (v:v) complexes of 2 antigens which can be identified from the row and column labels.

OBJECTIVE AND METHODS

Objective

In this analysis, we assessed the presence of autoantibodies against glycolipids, including gangliosides, at baseline in participants with CIDP in the ADHERE trial (NCT04281472)

Methods

- The methods of the ADHERE trial have been published[13]
- Serum samples collected at stage A baseline from 240 participants in ADHERE (excluding those from China) and 100 healthy controls were analyzed using a glycoarray multiplex assay (**Figure 1** and **Supplemental Figure S1**)
- Glycoarrays include 16 single glycolipid/phospholipid targets and 120 heteromeric complexes (1:1, V:V) printed in duplicate
- Fluorescently labeled anti-human IgG, IgM, and IgA antibodies quantified binding, measured in fluorescent intensity units
- Theoretical models of glycolipid presentation within plasma membrane due to cis-interactions are included in **Supplemental Figures S2** and **S3**

RESULTS

IgG- and IgA-Positive Signals in Patients With CIDP

- 62% versus 54% of baseline samples from patients with CIDP versus healthy controls showed an IgG-positive signal against an antigen exceeding average control reactivity threshold
- IgG binding was statistically significant against 6 heteromeric complexes containing GalNAc-GD1a, GM1, and/or LM1 in the CIDP samples (**Figure 2A**)
- Differential analysis identified the presence of both distinct and overlapping IgG binding patterns within individual participants in a subset of the CIDP cohort, and significantly elevated anti-glycolipid antibodies versus controls (**Figure 2B**)
- IgA binding patterns also differed, with reactivity against nine heteromeric complexes elevated in CIDP versus control samples, 67% (6/9) of which contained GalNAc-GD1a and/or GM2

IgM-Positive Signals in Patients With CIDP

- IgM reactivity was observed in both groups, with notable elevations in CIDP samples for 36 targets, particularly with GalNAc-GD1a and GM2 complexes (36%; 13/36) (**Figure 3**)

GalNAc-GD1a Glycolipid Profiles for IgG and IgM

- IgG and IgM reactivities against GalNAc-GD1a complexes were frequently found in CIDP patients. The most significant antibody reactivities are included in **Table 1**
- Anti-GalNAc-GD1a monospecific IgG localized to motor and sensory neurons, within compact myelin, paranodal, and periaxonal-axolemma regions[14]
- The CIDP participant population with significantly higher levels of IgM PS.GalNAc.GD1a were mostly negative for IgG of the same reactivity (**Figure 4**)

FIGURE 2 Heat map and Venn Diagram of IgG Anti-Glycolipid Autoantibodies

A. Heat map of the Significant Glycolipids for IgG

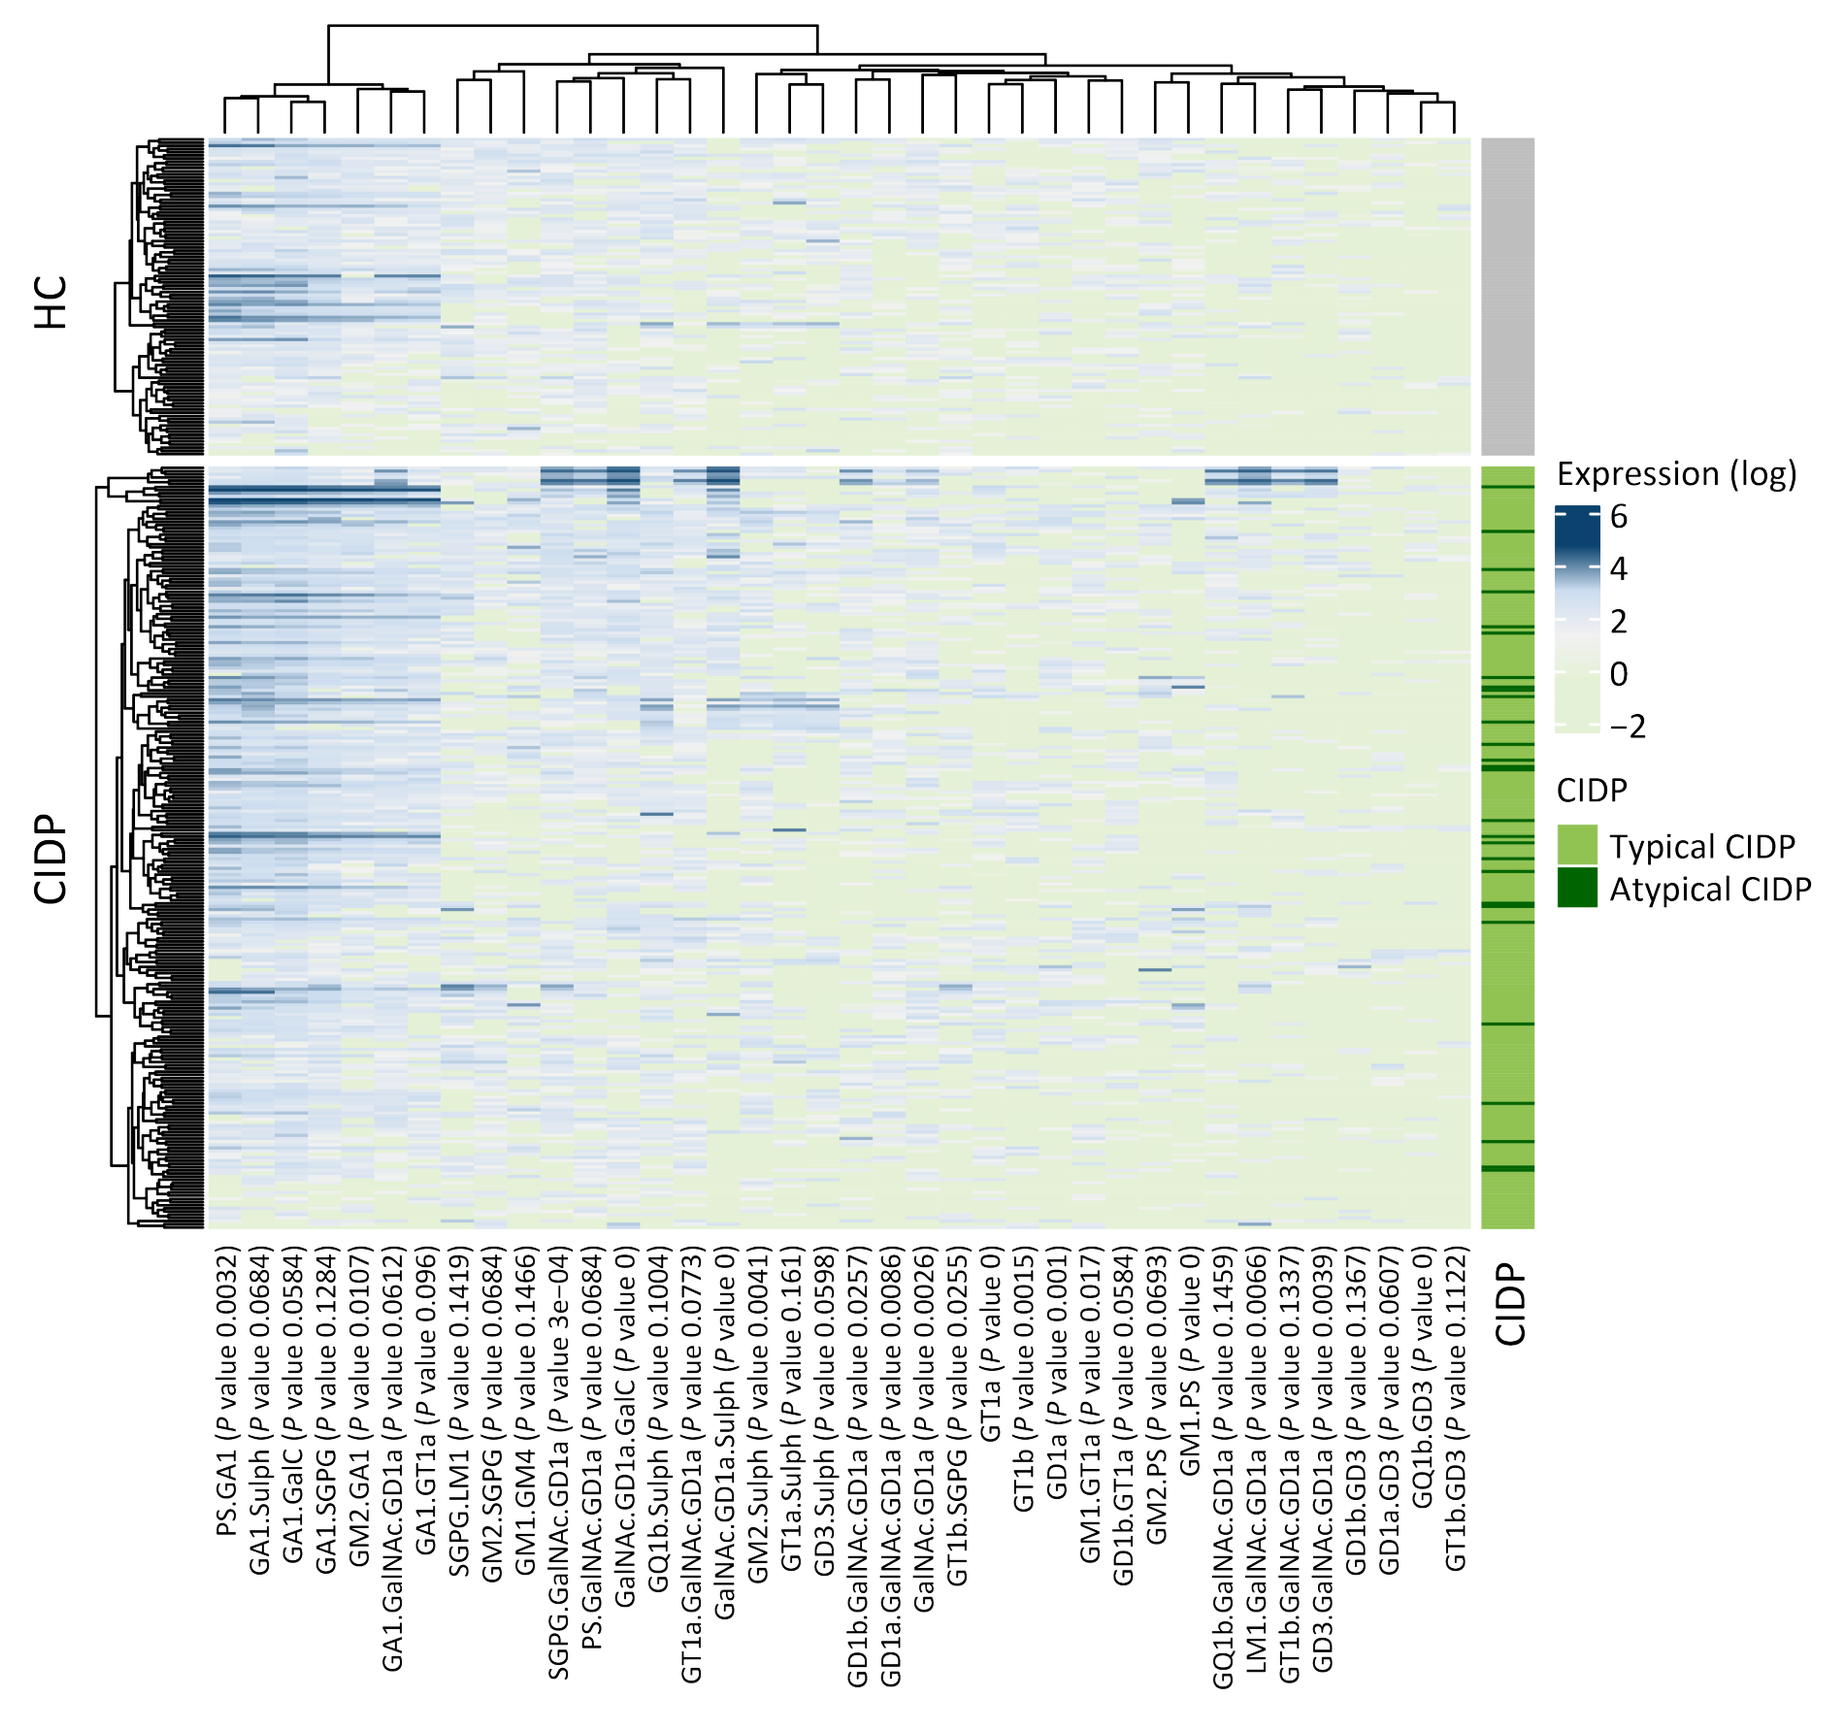
HC
CIDP
Expression (log)
6
4
2
0
−2
CIDP
Typical CIDP
Atypical CIDP
PS.GA1 (P value 0.0032)
GA1.Sulph (P value 0.0684)
GA1.GalC (P value 0.0584)
GA1.SGPG (P value 0.1284)
GM2.GA1 (P value 0.0107)
GA1.GalNAc.GD1a (P value 0.0612)
GA1.GT1a (P value 0.096)
SGPG.LM1 (P value 0.1419)
GM2.SGPG (P value 0.0684)
GM1.GM4 (P value 0.1466)
SGPG.GalNAc.GD1a (P value 3e−04)
PS.GalNAc.GD1a (P value 0.0684)
GalNAc.GD1a.GalC (P value 0)
GQ1b.Sulph (P value 0.1004)
GT1a.GalNAc.GD1a (P value 0.0773)
GalNAc.GD1a.Sulph (P value 0)
GM2.Sulph (P value 0.0041)
GT1a.Sulph (P value 0.161)
GD3.Sulph (P value 0.0598)
GD1b.GalNAc.GD1a (P value 0.0257)
GD1a.GalNAc.GD1a (P value 0.0086)
GalNAc.GD1a (P value 0.0026)
GT1b.SGPG (P value 0.0255)
GT1a (P value 0)
GT1b (P value 0.0015)
GD1a (P value 0.001)
GM1.GT1a (P value 0.017)
GD1b.GT1a (P value 0.0584)
GM2.PS (P value 0.0693)
GM1.PS (P value 0)
GQ1b.GalNAc.GD1a (P value 0.1459)
LM1.GalNAc.GD1a (P value 0.0066)
GT1b.GalNAc.GD1a (P value 0.1337)
GD3.GalNAc.GD1a (P value 0.0039)
GD1b.GD3 (P value 0.1367)
GD1a.GD3 (P value 0.0607)
GQ1b.GD3 (P value 0)
GT1b.GD3 (P value 0.1122)
CIDP

B. Differential Analysis of Distinct and Overlapping IgG Binding Patterns

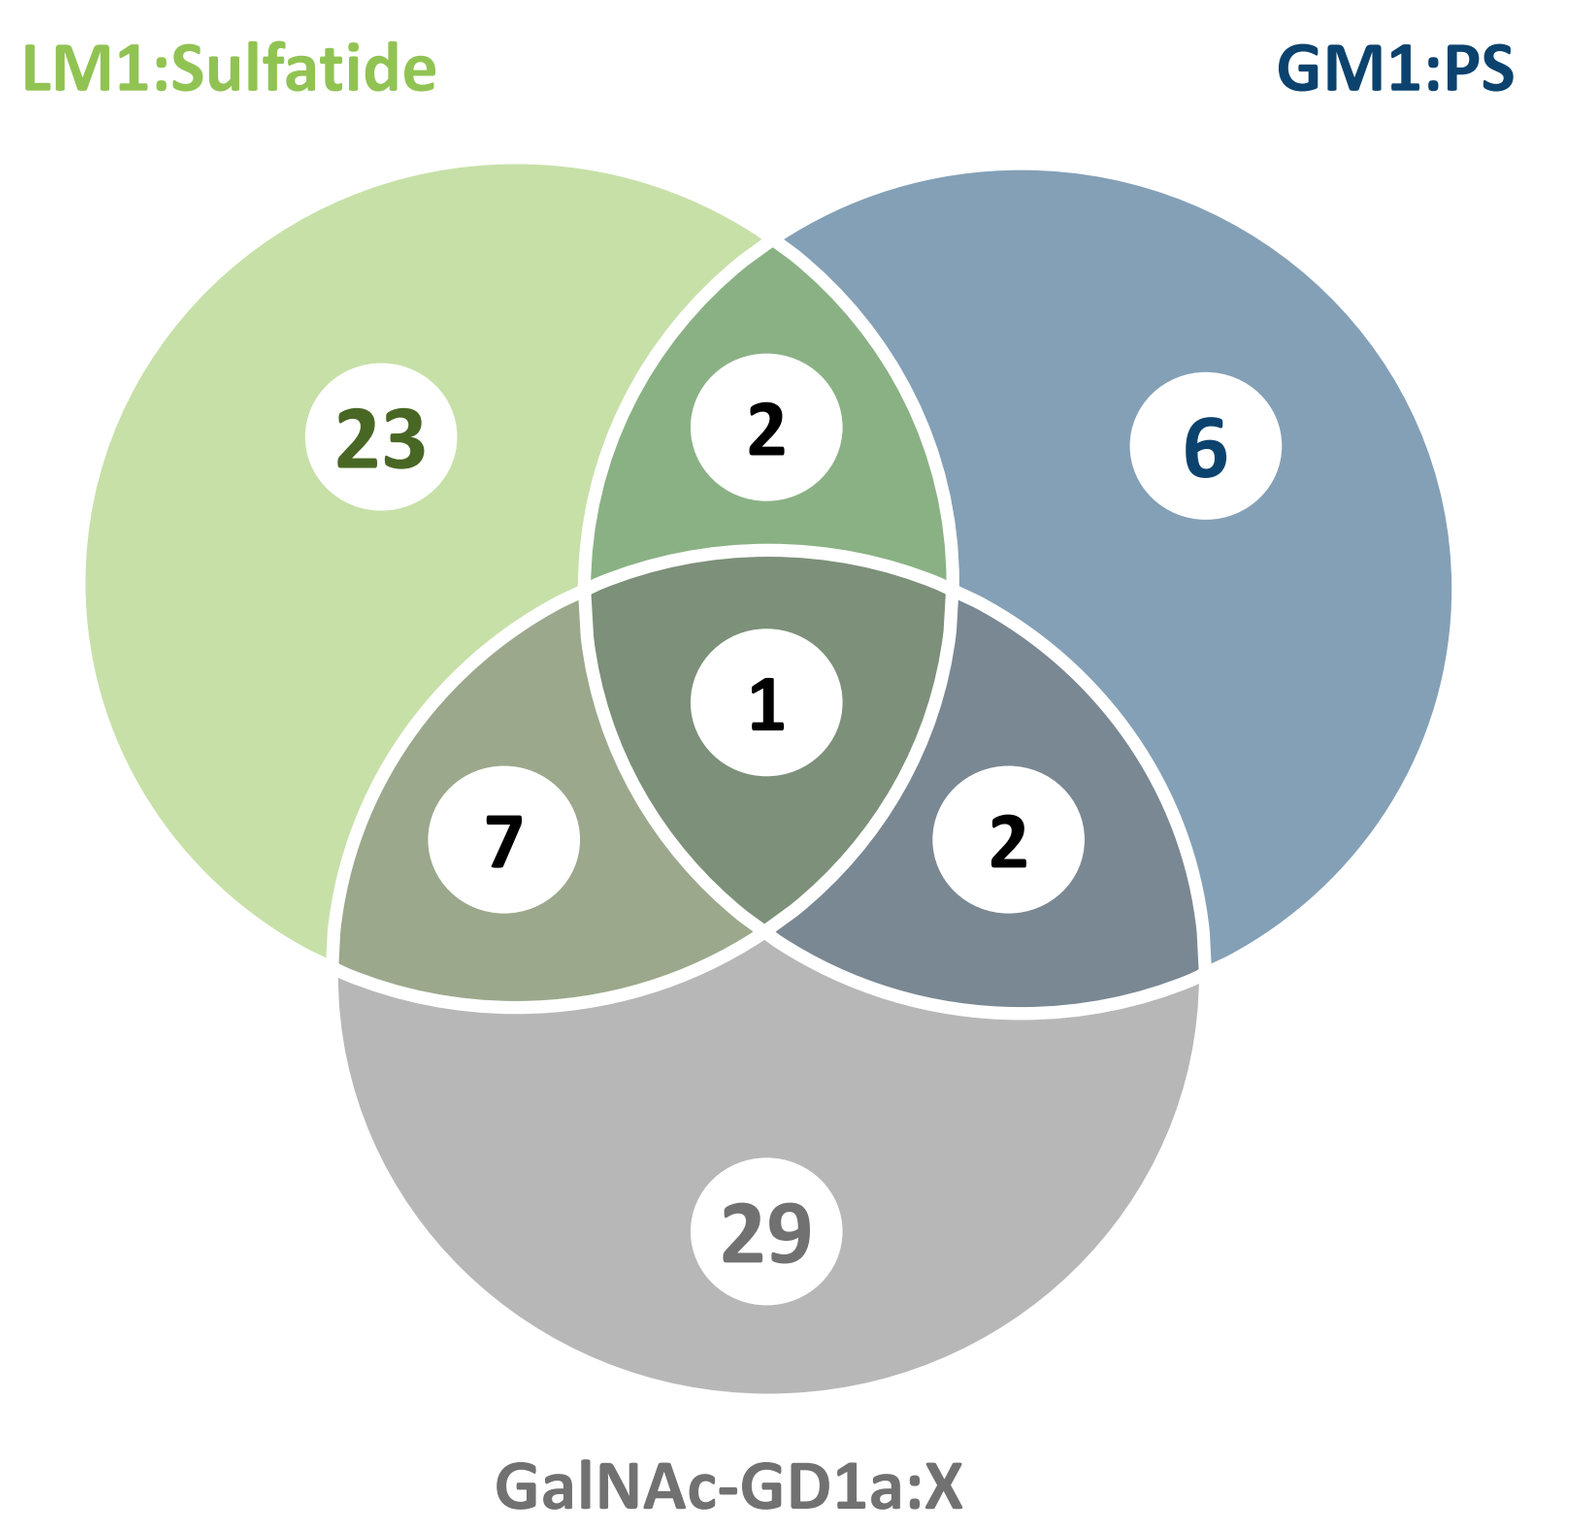
LM1:Sulfatide
GM1:PS
23
2
6
1
7
2
29
GalNAc-GD1a:X

A. Heat map displaying IgG anti-glycolipid/phospholipid binding patterns in a cohort of CIDP and HCs. Columns represent antigen targets and rows represent each patient or control. A diverging scale is utilized to demonstrate FIU (log 10) of bound IgG per target per patient, whereby no binding appears light green, weak binding is blue, and strongest binding is indicated as dark blue. The adjusted *P* value of each target is represented. Typical and atypical CIDP are included on the right-hand side of the heat map. Typical CIDP[2] (symmetric, proximal, and distal muscle weakness of upper and lower limbs, either progressive or relapsing, and with sensory involvement in two limbs). Atypical CIDP[2] (distal [predominantly in lower limbs], multifocal [usually asymmetric, upper limb predominant], or focal [only one limb] sensory loss and muscle weakness, or motor [without sensory involvement]). **The adjusted *P* value is attached to the glycolipid name.**

B. Venn diagram highlighting the presence of both distinct and overlapping IgG binding patterns within individual patients in a subset of the CIDP cohort. Each patient had elevated IgG reactivity to one or more of the 3 significant antigen targets LM1:Sulfatide, GM1:Phosphatidylserine (PS), and/or GalNAc-GD1a: X (X= Sulfatide, GalC, PS, and/or LM1 complexes).

FIGURE 3 Heat map of IgM Anti-Glycolipid Autoantibodies

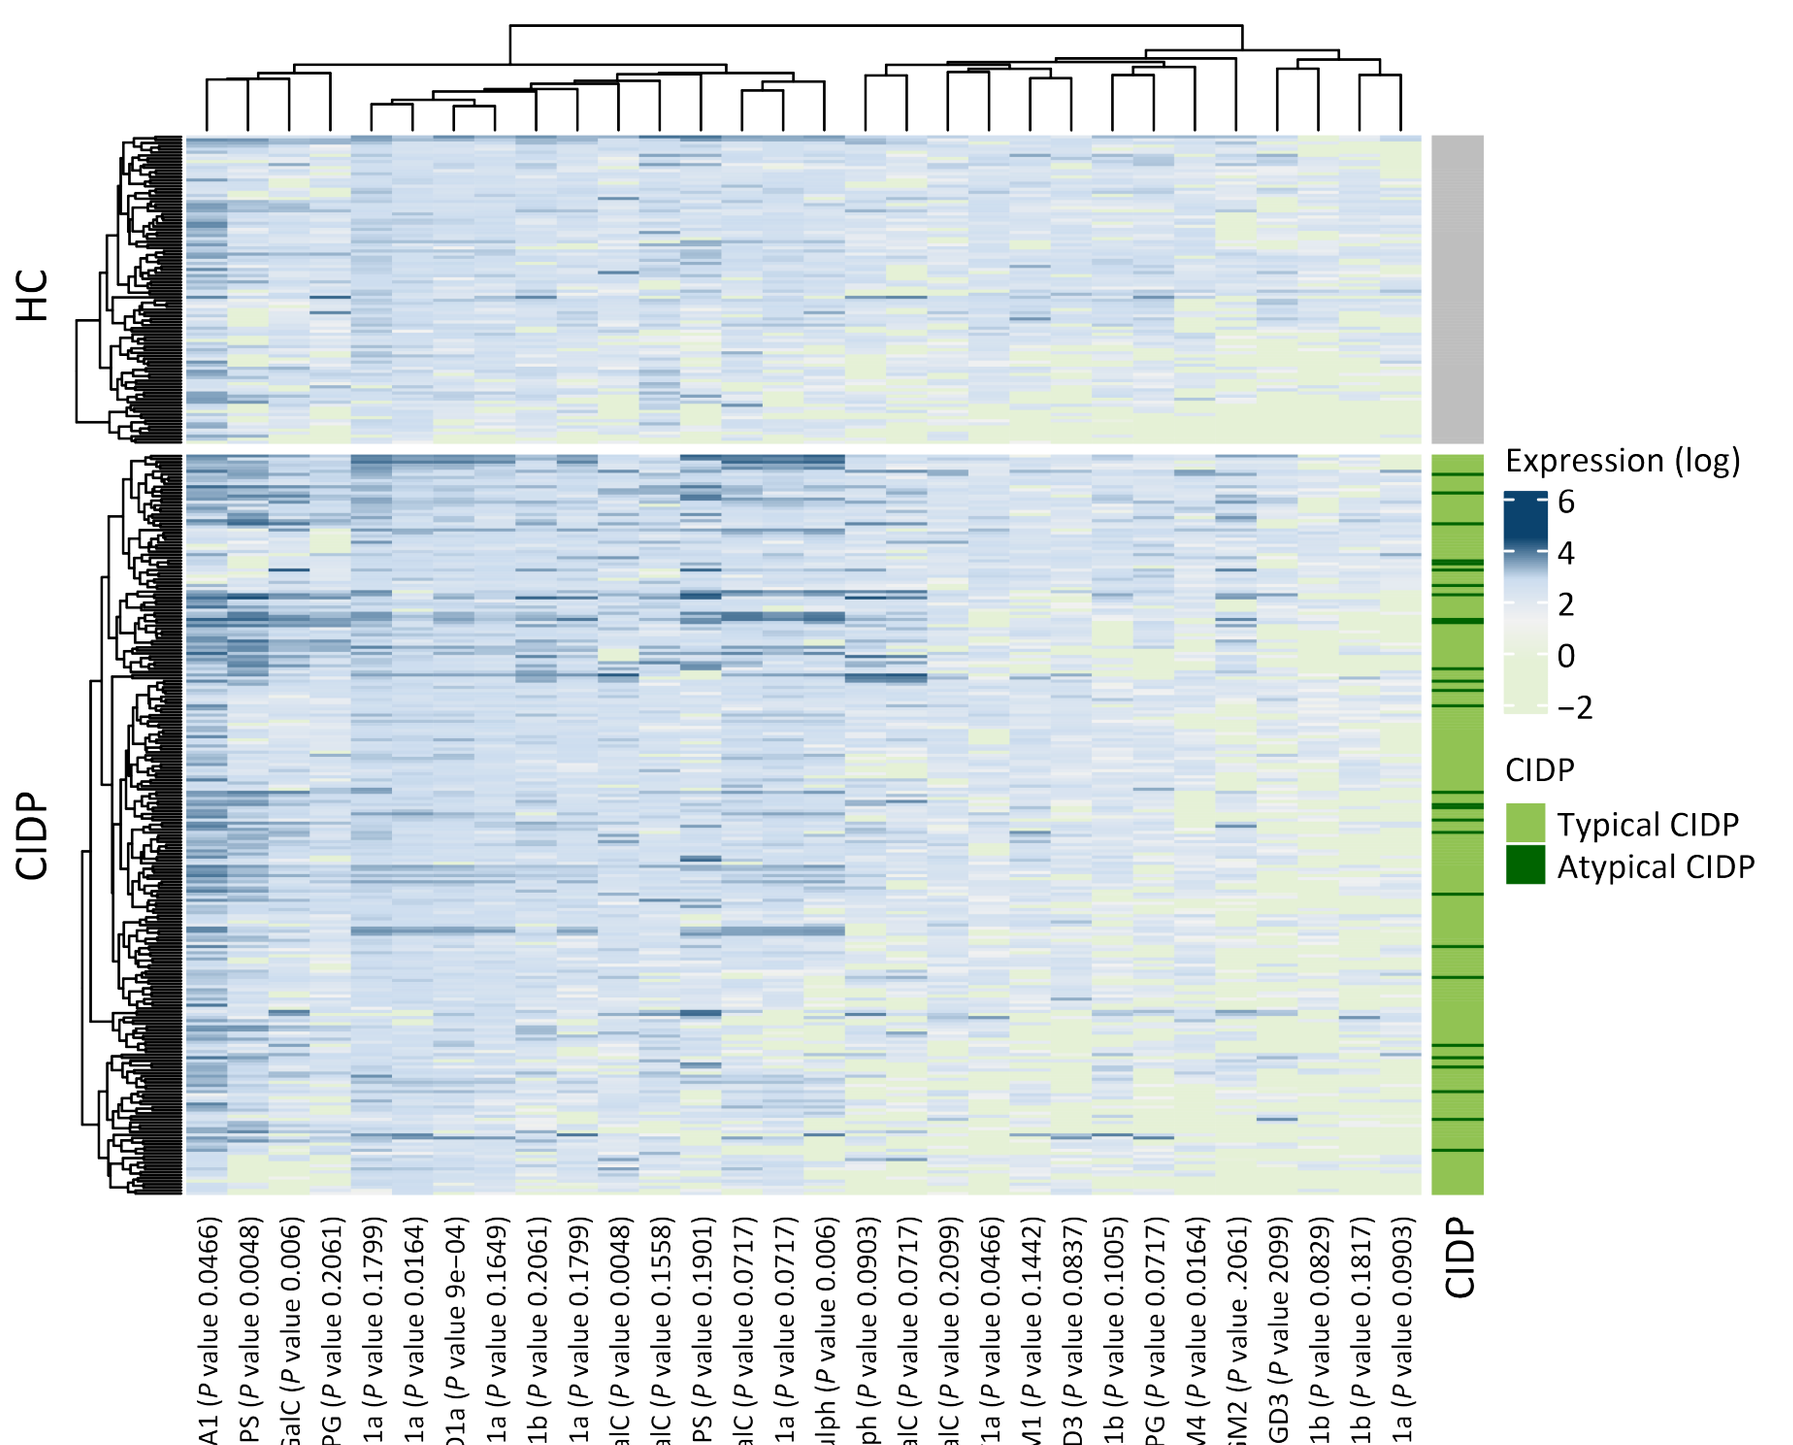
HC
CIDP
Expression (log)
6
4
2
0
−2
CIDP
Typical CIDP
Atypical CIDP
GM2.GA1 (P value 0.0466)
GM1.PS (P value 0.0048)
GM1.GalC (P value 0.006)
GM1.SGPG (P value 0.2061)
SGPG.GalNAc.GD1a (P value 0.1799)
GT1a.GalNAc.GD1a (P value 0.0164)
PS.GalNAc.GD1a (P value 9e−04)
GM4.GalNAc.GD1a (P value 0.1649)
PS.GD1b (P value 0.2061)
GD1b.GalNAc.GD1a (P value 0.1799)
GT1b.GalC (P value 0.0048)
PS.GalC (P value 0.1558)
GM2.PS (P value 0.1901)
GalNAc.GD1a.GalC (P value 0.0717)
LM1.GalNAc.GD1a (P value 0.0717)
GalNAc.GD1a.Sulph (P value 0.006)
GD1b.Sulph (P value 0.0903)
GD1b.GalC (P value 0.0717)
GD3.GalC (P value 0.2099)
GT1a (P value 0.0466)
GD3.LM1 (P value 0.1442)
GT1b.GD3 (P value 0.0837)
GM2.GQ1b (P value 0.1005)
GD1a.SGPG (P value 0.0717)
GM4 (P value 0.0164)
GM1.GM2 (P value .2061)
GM2.GD3 (P value 2099)
GD1a.GT1b (P value 0.0829)
GT1b (P value 0.1817)
GD1a (P value 0.0903)
CIDP

Heat map displaying IgM anti-glycolipid/phospholipid binding patterns in a cohort of CIDP and HCs. Columns represent antigen targets and rows represent each patient or control. A diverging scale is utilized to demonstrate FIU of bound IgM per target per patient, whereby no binding appears light green, weak binding is blue, and strongest binding is indicated as dark blue. The adjusted *P* value of each target is represented. Typical and atypical CIDP are included on the right-hand side of the heat map. Typical CIDP[2] (symmetric, proximal, and distal muscle weakness of upper and lower limbs, either progressive or relapsing, and with sensory involvement in two limbs). Atypical CIDP[2] (distal [predominantly in lower limbs], multifocal [usually asymmetric, upper limb predominant] or focal [only one limb] sensory loss and muscle weakness, or motor [without sensory involvement]).

The adjusted *P* value is attached to the glycolipid name.

TABLE 1 Most Significant IgM and IgG With Reactivities Against GalNAc-GD1a Complexes Resulting From Weighted Linear Modeling*

Antibody	Symbol	logFC	*P* value	Adjusted *P* value
IgG	GalNAc-GD1a:GalC	0.4291	0.0000	0.0000
IgG	GalNAc-GD1a:Sulph	0.5133	0.0000	0.0000
IgG	SGPG:GalNAc-GD1a	0.2921	0.0000	0.0003
IgG	GD3:GalNAc-GD1a	0.3778	0.0003	0.0039
IgG	GalNAc-GD1a	0.2401	0.0002	0.0026
IgG	LM1:GalNAc-GD1a	0.3413	0.0006	0.0066
IgG	GD1a:GalNAc-GD1a	0.2058	0.0009	0.0086
IgM	PS:GalNAc-GD1a	0.2015	0.0000	0.0009
IgM	GalNAc-GD1a:Sulph	0.2239	0.0002	0.0060
IgM	GT1a:GalNAc-GD1a	0.1265	0.0008	0.0164
IgM	GalNAc-GD1a:GalC	0.1549	0.0054	0.0717
IgM	LM1:GalNAc-GD1a	0.1413	0.0066	0.0717
IgM	GM4:GalNAc-GD1a	0.0966	0.0255	0.1649

*Log10 fold changes, p-values, adjusted p-values (FDR), and 95% CI are shown. Weights were incorporated using a sigmoid-transformed covariate (midpoint = 3, slope = 0.005), resulting in weights approaching 1 when the binding value (on a log10 scale) exceeds 1000. Linear models were fitted using limma package in R, with empirical Bayes moderation and contrast-based inference.

FIGURE 4 Representative Distribution of IgG and IgM of the Same Reactivity Shows a Distinct Signature for GalNAc.GD1a.GalC and GalNAc.GD1a.PS

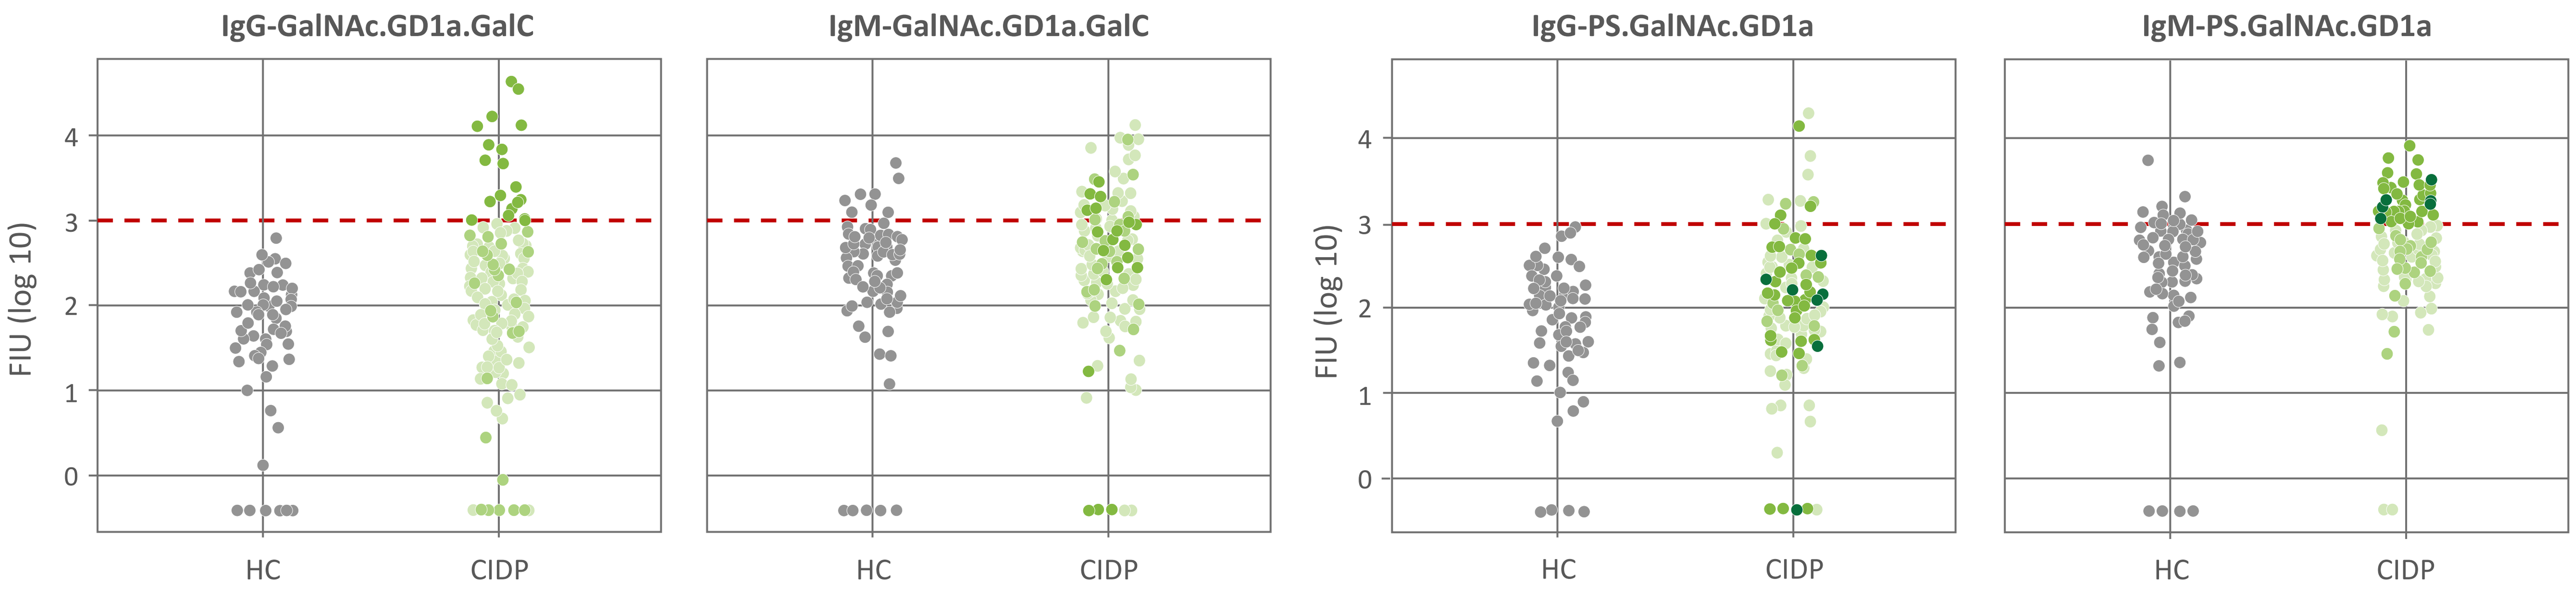
IgG-GalNAc.GD1a.GalC
IgM-GalNAc.GD1a.GalC
IgG-PS.GalNAc.GD1a
IgM-PS.GalNAc.GD1a
FIU (log 10)
4
3
2
1
0
HC
CIDP

Dotted red line indicates negative signal cutoff. Each colored dot in the 2 graphs represents the same CIDP patient population. Light green represents typical CIDP and dark green dots represent atypical CIDP. Bright colors are selected for the population of interest. Typical CIDP[2] (symmetric, proximal, and distal muscle weakness of upper and lower limbs, either progressive or relapsing, and with sensory involvement in 2 limbs). Atypical CIDP[2] (distal [predominantly in lower limbs], multifocal [usually asymmetric, upper limb predominant] or focal [only one limb] sensory loss and muscle weakness, or motor [without sensory involvement]).

KEY TAKEAWAYS

Translational data for IgG in the ADHERE trial identified LM1 complexes and GalNAc-GD1a complexes as glycolipid targets of interest in CIDP

Translational data for IgM identified specific targets in CIDP samples, notably GalNAc-GD1a and GM2 complexes

CIDP-specific anti-glycolipid IgG signatures suggest a potential role for these antigens in CIDP pathology and as biomarkers

These results highlight a successful experimental approach using individual and heteromeric complexes for glycolipid/phospholipid antigen targets in CIDP. Additional studies may reveal whether the IgG and IgM antibodies identified can provide insight into the pathogenesis, diagnosis, and prognosis of CIDP

Presented at the 2025 Peripheral Nerve Society (PNS) Annual Meeting; May 17–20, 2025; Edinburgh, UK

ABBREVIATIONS

CI, confidence interval; CIDP, chronic inflammatory demyelinating polyradiculoneuropathy; FDR, false discovery rate; FIU, fluorescence intensity units; GalC, galactosylceramide; GalNAc, N-acetylgalactosamine; GD, disialoganglioside; GM, monosialoganglioside; GT1a, GT trisialoganglioside; HC, healthy control; Ig, immunoglobulin; IVIg, intravenous immunoglobulin; LM1, monosialylated lactosylceramide; logFC, log fold change; PS, phosphatidylserine; SC, subcutaneous; SGPG, sulfoglucuronosyl paragloboside.

DISCLOSURES AND ACKNOWLEDGMENTS

SKH: Funding: argenx to conduct this project; **HJW:** Consultancy: argenx; **AR, EH, GI, BB:** Employees: argenx.

Preliminary analysis of the differential expression of glycolipids between CIDP and healthy controls for IgG was performed by Yingjie Zhang.

REFERENCES

1. Cox SZ, Gwathmey KG. *Clin Geriatr Med*. 2021;37:327–45.
2. Van den Bergh PYK, et al. *Eur J Neurol*. 2021;28:3556–83.
3. Querol L, et al. *Sci Rep*. 2017;7:14411.
4. Mathey EK, et al. *J Neurol Neurosurg Psychiatry*. 2015;86:973–85.
5. Yan WX, et al. *Ann Neurol*. 2000;47:765–75.
6. Manso C, et al. *J Clin Invest*. 2019;129:2222–36.
7. Querol L, Lleixà C. *Neurotherapeutics.* 2021;18:2222–35.
8. van Doorn IN, et al. *Ther Clin Risk Manag.* 2024;20:111–126.
9. Collet R, et al. *Rev Neurol (Paris).* 2023;179(8):831–843.
10. Kister A, Kister I. *Front Chem.* 2023;10:1041961.
11. Rinaldi S, et al. *PLoS ONE* 2013;8:e82337.
12. Ulrichts P, et al. *J Clin Invest.* 2018;128:4372–86.
13. Allen JA, et al. *Lancet Neurol.* 2024;23:1013–1024.
14. Kaida K, et al. *Neurology.* 2003;61:465–70.

SCAN ME