174

Assessing Efgartigimod Dosing Patterns and Myasthenia Gravis Activities of Daily Living Outcomes in Clinical Practice: Results from a Large Patient Support Program Database in the United States

Pushpa Narayanaswami[1], Martina Orlovic[2], Cynthia Qi[2], Ratna Bhavaraju-Sanka[3], A. Gordon Smith[4], Matthew Jefferson[2], Edward Brauer[2], Deborah Gelinas[2], Rohit R Menon[5], Mai Sato[6], Gil I. Wolfe[7]

[1]Department of Neurology, Beth Israel Deaconess Medical Center, 330 Brookline Avenue, Boston, MA, USA; [2]argenx US Inc., 33 Arch Street, 32nd Floor, Boston, MA, USA; [3]Department of Neurology, University of Texas Health Science Center at San Antonio, San Antonio, TX, USA; [4]Department of Neurology, Virginia Commonwealth University, 1101 E Marshall Street, Richmond, VA, USA; [5]ZS Associates, MFAR Manyata Tech Park, 10th Cross Road, Bengaluru, Karnataka, India; [6]ZS Associates, 350 Fifth Avenue, New York, NY, USA; [7]Department of Neurology, Jacobs School of Medicine and Biomedical Sciences, University at Buffalo, 1010 Main Street, Buffalo, NY, USA.

INTRODUCTION

- Efgartigimod is an engineered human Fc-fragment that is approved in the United States, European Union, Canada, and China for treatment of anti-acetylcholinesterase receptor antibody positive generalized myasthenia gravis (gMG), and gMG regardless of antibody status in Japan.[1-6]
 - **Dosing guidance based on label:** Each efgartigimod cycle consists of 1 infusion each week for 4 weeks; intervals between cycles vary based on individual response, enabling adaptability for heterogeneous phenotypes.[1]
 - **Expected outcomes based on trial:** In ADAPT, a significantly greater proportion of efgartigimod-treated patients achieved clinically meaningful improvement (CMI; ≥2-point change) in Myasthenia Gravis Activities of Daily Living (MG-ADL) at week 4 (77.8% vs. 48.3%).[2]
- There is a large interest in evaluating treatment cycle patterns and outcomes associated with efgartigimod in routine clinical practice. While broader population-level evidence is limited, several single-center studies and case studies have reported early results consistent with expectations:
 - **Treatment cycle patterns observed in clinical practice:** Across several studies, most patients received the first 2 to 3 treatment cycles with intervals of 4 weeks between each cycle, with intervals adjusted in subsequent cycles according to individual clinical response.[7-9] Other studies have also reported on more flexible approaches from earlier cycles.[4,10]
 - **Outcomes observed in clinical practice:** Across studies, CMI was achieved by the majority of patients, with the mean first best follow-up MG-ADL in the range of 4 to 6 points improvement from baseline commonly within the first 6 months of treatment.[7-10]
- The objective of this study was to evaluate dosing patterns and MG-ADL outcomes associated with a broad spectrum of patients with gMG using efgartigimod in the US, to augment existing evidence. We utilized the largest database of efgartigimod initiators to date, from the My VYVGART Path patient support program (PSP), up to April 1, 2025.

METHODS

- **Dataset:** Data were obtained from the My VYVGART Path PSP, which provides personalized Nurse Case Manager support for enrolled patients with gMG. Through regular phone contact, the PSP has been capturing patient baseline characteristics (e.g., age, gender), efgartigimod dosing information (e.g., infusion dates), and MG-ADL scores from those enrolled since January 2022 after efgartigimod launch in the US. The PSP data were also integrated with dispense and MG-ADL data from specialty pharmacies.
- **Cohort selection and outcomes:** Patients with gMG enrolled in the PSP (aged ≥18 years by default) who had initiated efgartigimod by April 1, 2025, were screened. Those who had a baseline MG-ADL available, and at least 4 MG-ADL scores post-efgartigimod initiation captured during their available follow-up, were included. Outcomes were assessed for the maximum follow-up available.

Efgartigimod dosing patterns

- **Treatment cycles** were defined as time from the first to last efgartigimod infusion administered 1 week apart.
- **Gaps** between cycles were defined as last infusion of the previous cycle to the first infusion of the subsequent cycle.
- All periods were defined as weeks, from 0 weeks (0 to <7 days), 1 week (7 to <14 days), etc.

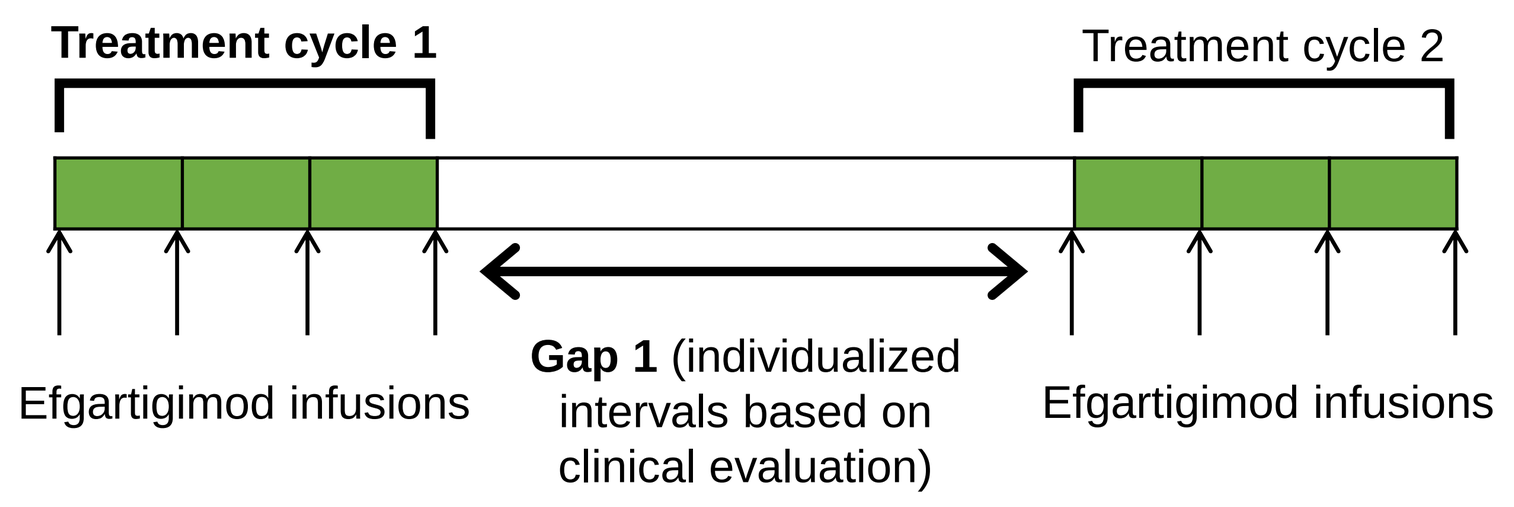

Treatment cycle 1
Treatment cycle 2
Efgartigimod infusions
Gap 1 (individualized intervals based on clinical evaluation)
Efgartigimod infusions

MG-ADL outcomes

- **Largest observed response (best state):** Difference between the lowest observed MG-ADL after efgartigimod initiation at any time vs. baseline (n=2648 data points for each patient).
- **Correlation with time on treatment:** To assess outcomes by efgartigimod treatment cycle, any patients with at least 1 MG-ADL score captured within each cycle were included. For outcomes by month, any patients with at least 1 MG-ADL score captured in any given month were included. If multiple scores were captured during a given cycle or month, only the best (lowest) score was considered. Variation in patients with scores available across cycles and months are denoted in the figures.

The above outcomes were designed to provide population-level insights given the heterogeneous timing of MG-ADL capture among all patients:

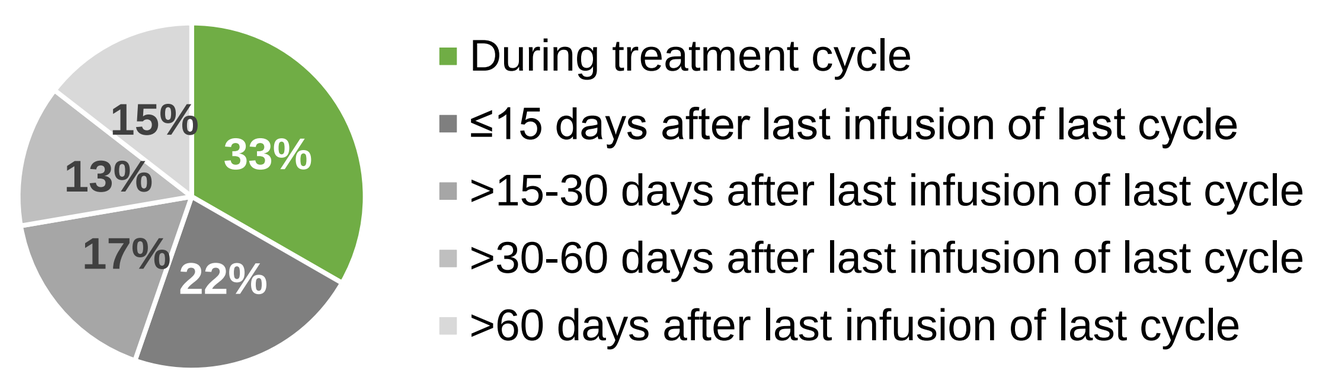

33%
22%
17%
13%
15%
During treatment cycle
≤15 days after last infusion of last cycle
>15-30 days after last infusion of last cycle
>30-60 days after last infusion of last cycle
>60 days after last infusion of last cycle

SUMMARY

Utilizing the largest available dataset (N=2648 patients) among novel therapies in gMG, our analysis represents the most direct and up-to-date analysis of dosing patterns and outcomes associated with efgartigimod in US clinical practice.

- Despite substantial symptom burden at baseline (84% with MG-ADL≥5; 36% with MG-ADL≥10), 83% patients achieved a mild disease state of MG-ADL ≤4 after efgartigimod.
- Substantial MG-ADL reduction (~4 points) was observed as early as Month 1 and sustained up to Month 12 after efgartigimod treatment initiation; the magnitude of benefit was consistent across treatment cycles and over time.
- Efgartigimod treatment cycles were individualized based on clinical evaluation, with 4 weeks being the most common gap length between cycles.

Some limitations should be noted: 1) the study cohort was limited to patients enrolled in the PSP who had initiated efgartigimod by April 1, 2025; 2) the MG-ADL data analyzed were self-reported by patients, with limited consistency in timepoint and frequency of capture; 3) no safety data or concomitant medication information were collected as part of the dataset; 4) the findings reflect observation in clinical practice where external factors (e.g., payer policies) could influence utilization patterns and outcomes.

As dosing patterns and outcomes associated with efgartigimod may continue to evolve, further analyses should be conducted to provide more insights to help optimize outcomes for patients.

RESULTS

Among 2648 efgartigimod initiators with gMG, gaps between treatment were most commonly 4 weeks

- 2648 (38%) of 6967 efgartigimod initiators with robust MG-ADL capture were included (Figure 1). Baseline patient characteristics were consistent with expectations, with heavier representation of government insurance (Table 1).
- Most patients (95%) had initiated at least Cycle 2 of efgartigimod treatment (mean=6.3 cycles). When evaluated week-by-week, gaps were most commonly 4 weeks, with over half of all gaps being 4-6 weeks (Figure 2).

Figure 1. Patient selection

Patients with gMG enrolled in the PSP who initiated efgartigimod by April 1, 2025
n=6967[a] (100%)

→ Baseline and at least 1 follow-up MG-ADL score available
n=4277 (61%)

→ At least 4 follow-up MG-ADL scores available
n=2648 (38%)

[a]Enrolled patients with negative values or confounding records for efgartigimod infusion-related data were excluded from the study. To provide a comprehensive representation, useable data from any patients who had discontinued efgartigimod were included in the analysis.

Table 1. Baseline demographics and characteristics

	N=2648
Age	
Mean (SD), years	67.5 (15)
Distribution, n (%)	
18 to <50 years	341 (13)
50 to <65 years	549 (21)
≥65 years	1758 (66)
Sex, n (%)	
Men	1491 (56)
Women	1132 (43)
Unknown	25 (1)
Insurance, n (%)[a]	
Commercial	792 (30)
Government	1899 (72)
Other	105 (4)
Baseline MG-ADL (before efgartigimod)	
Mean (SD), points	8.2 (3.7)
Median (IQR), points	8 (6-11)
Range, points	0-22
Distribution, n (%)	
0-4 points	424 (16)
5-9 points	1259 (48)
10-24 points	965 (36)
Prior gMG treatments (before efgartigimod), n (%)[b]	
Information available	2047 (77)
AChE inhibitors	823 (40)
Any NSIST	569 (28)
Any GC	1285 (63)
Any advanced therapy[c]	1315 (64)

[a]Percentages may not add up to 100 as patients can be tagged to multiple payers. "Other" included patients tagged to Patient Assistance Programs (PAP) or patients who have no payer information available. [b]Percentages may not add up to 100 as patients can report multiple classes of treatments. [c]"Advanced therapy" included eculizumab, rituximab, ravulizumab, intravenous or subcutaneous immunoglobulin (IVIG), and/or plasma exchange (PLEX).

Figure 2. Efgartigimod dosing patterns

	N=2648
Follow-up since efgartigimod initiation	
Mean, weeks	89
Range, weeks	6-165
Efgartigimod formulation used, n (%)	
Intravenous (IV) only	1994 (75)
Subcutaneous (SC) only	241 (9)
Both IV and SC	413 (16)
Efgartigimod cycles initiated	
Mean, cycles	6.3
Range, cycles	1-28
Initiated at least Cycle 2, n (%)	2521 (95)
Initiated at least Cycle 6, n (%)	1293 (49)
Distinct gaps identified	
Number of gaps	13924

Distribution of all measurable gap lengths between treatment cycles (13,924 gaps)[a]

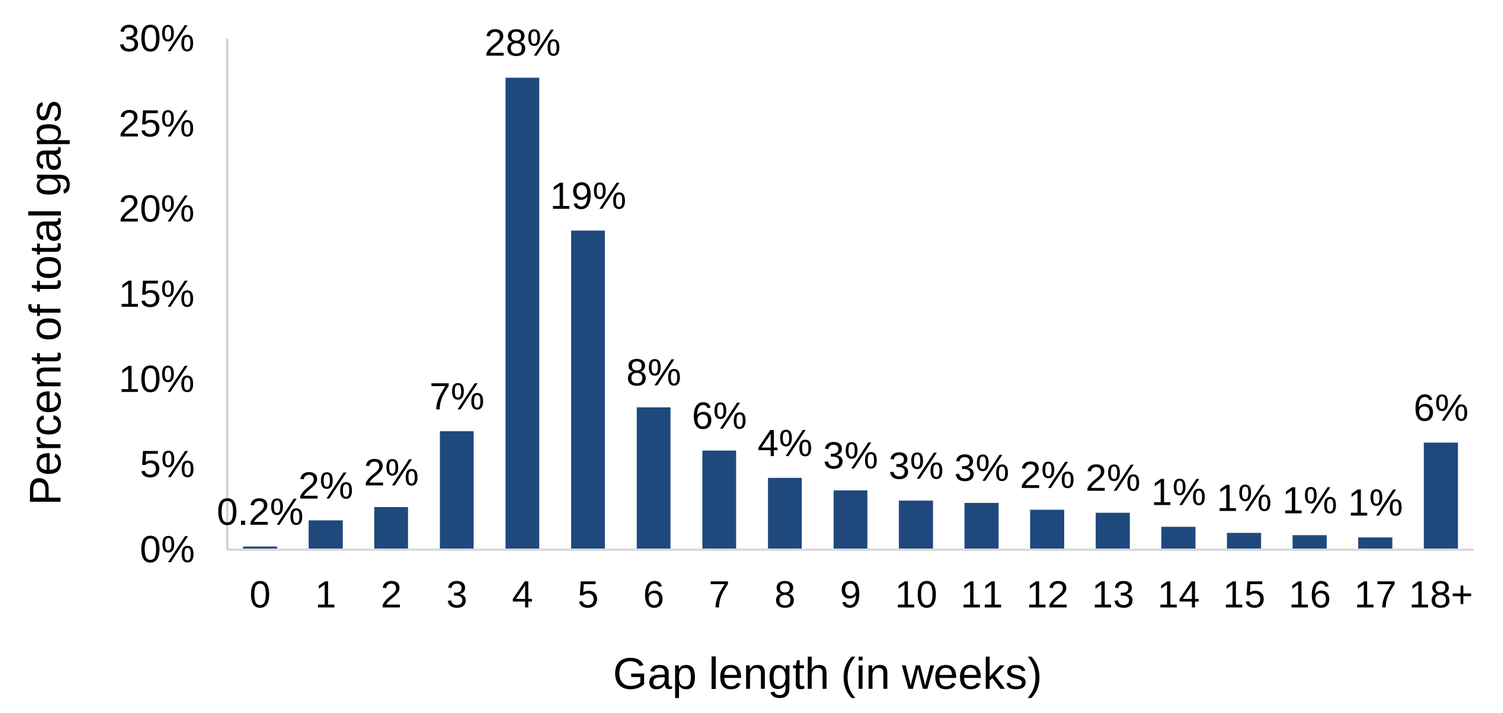

Percent of total gaps
30%
25%
20%
15%
10%
5%
0%
0.2% 2% 2% 7% 28% 19% 8% 6% 4% 3% 3% 3% 2% 2% 1% 1% 1% 1% 6%
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18+
Gap length (in weeks)

[a]Proportions were assessed weekly, with categories ranging from 0 to 18+ weeks. 0 weeks included gaps of 0 to <7 days, 1 week included gaps of 7 to <14 days, 2 weeks included gaps of 14 to <21 days, and so forth. Percentages were calculated against 13924 total measurable gaps of finite length identified among the 2648 patients.

The majority of patients experienced CMI after efgartigimod initiation, and in their best state, 83% achieved an MG-ADL score of 4 or lower

- Among largest observed responses at the patient-level, a mean of 5.8 points reduction from baseline was observed, with 91% of patients experiencing CMI (Figure 3).
- When all captured scores were considered for sensitivity analysis (24,223 total MG-ADL scores), the mean MG-ADL reduction was 3.8 points from baseline. The majority experienced CMI with efgartigimod regardless of age, sex, or baseline disease severity.
- At their best state after efgartigimod initiation, 83% of patients achieved MG-ADL scores of 4 or less, representing a state with mild disease (Figure 4).

Figure 3. Largest observed response after efgartigimod initiation

Largest observed response (N=2648[a])

▼ **5.8** points mean reduction from baseline, with **91%** of patients experiencing clinically meaningful improvement (≥2 points)

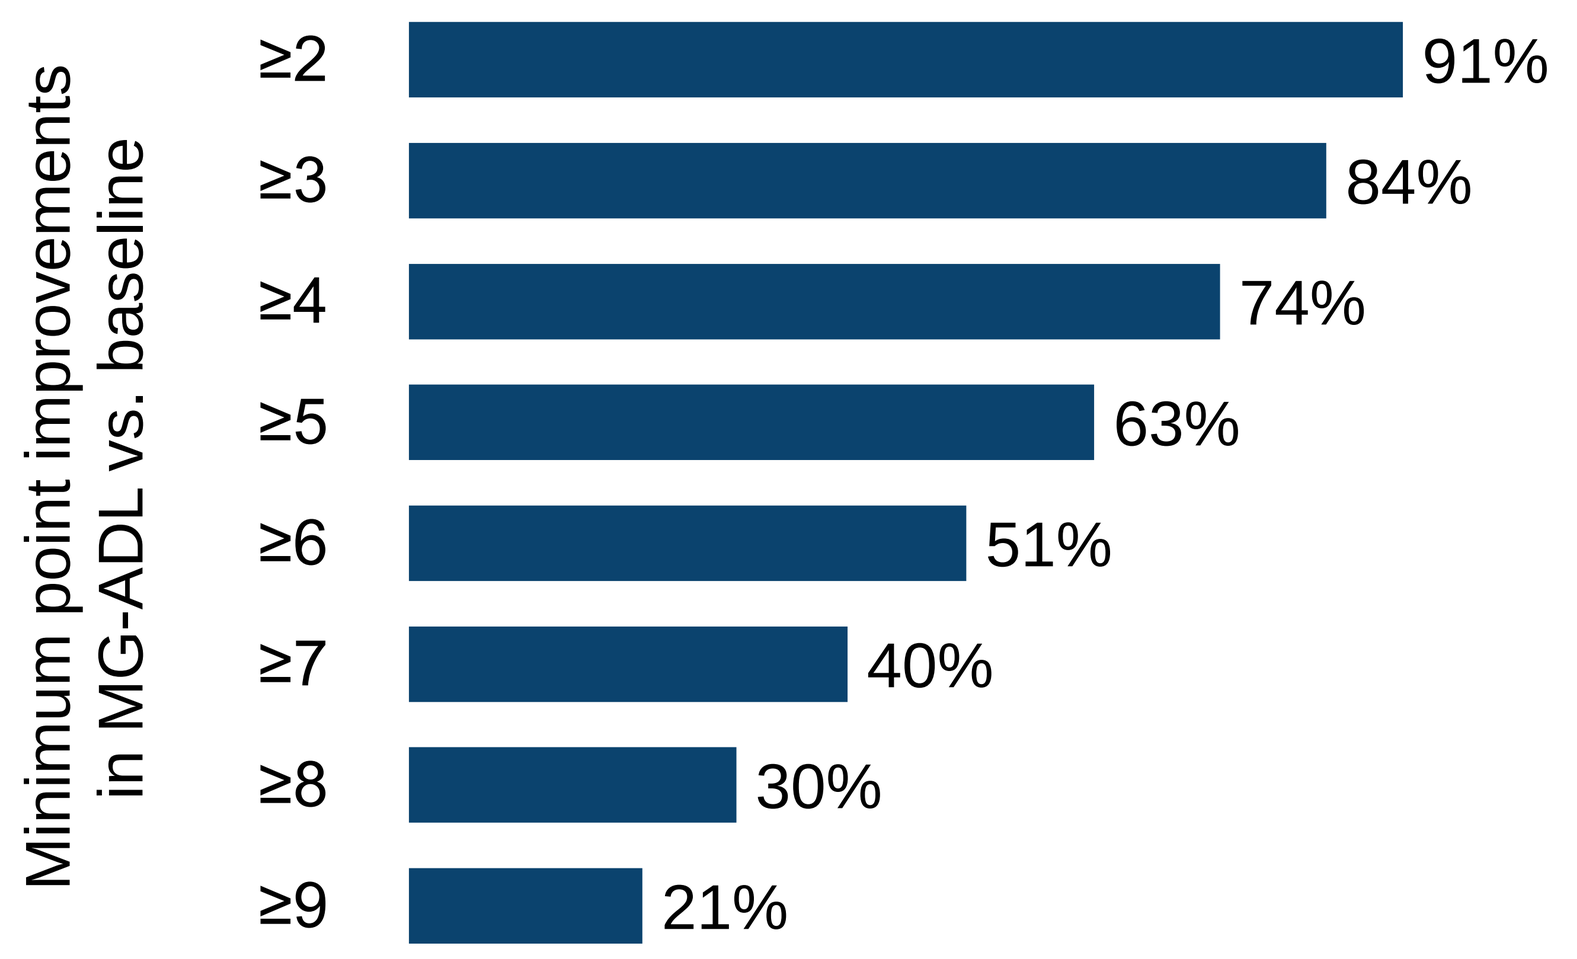

Minimum point improvements in MG-ADL vs. baseline
≥2 91%
≥3 84%
≥4 74%
≥5 63%
≥6 51%
≥7 40%
≥8 30%
≥9 21%

[a]Overall MG-ADL response after efgartigimod initiation across all timepoints captured was assessed in 2 ways: Largest observed response was calculated using the lowest MG-ADL score recorded by the patient any time after efgartigimod initiation and represents the best response. Average observed response was calculated using every captured MG-ADL record across all available timepoints to provide a full representation of the patient experience accounting for the fluctuations in MG-ADL during their treatment journeys.

Figure 4. Best state after efgartigimod initiation

Distribution of lowest observed MG-ADL scores

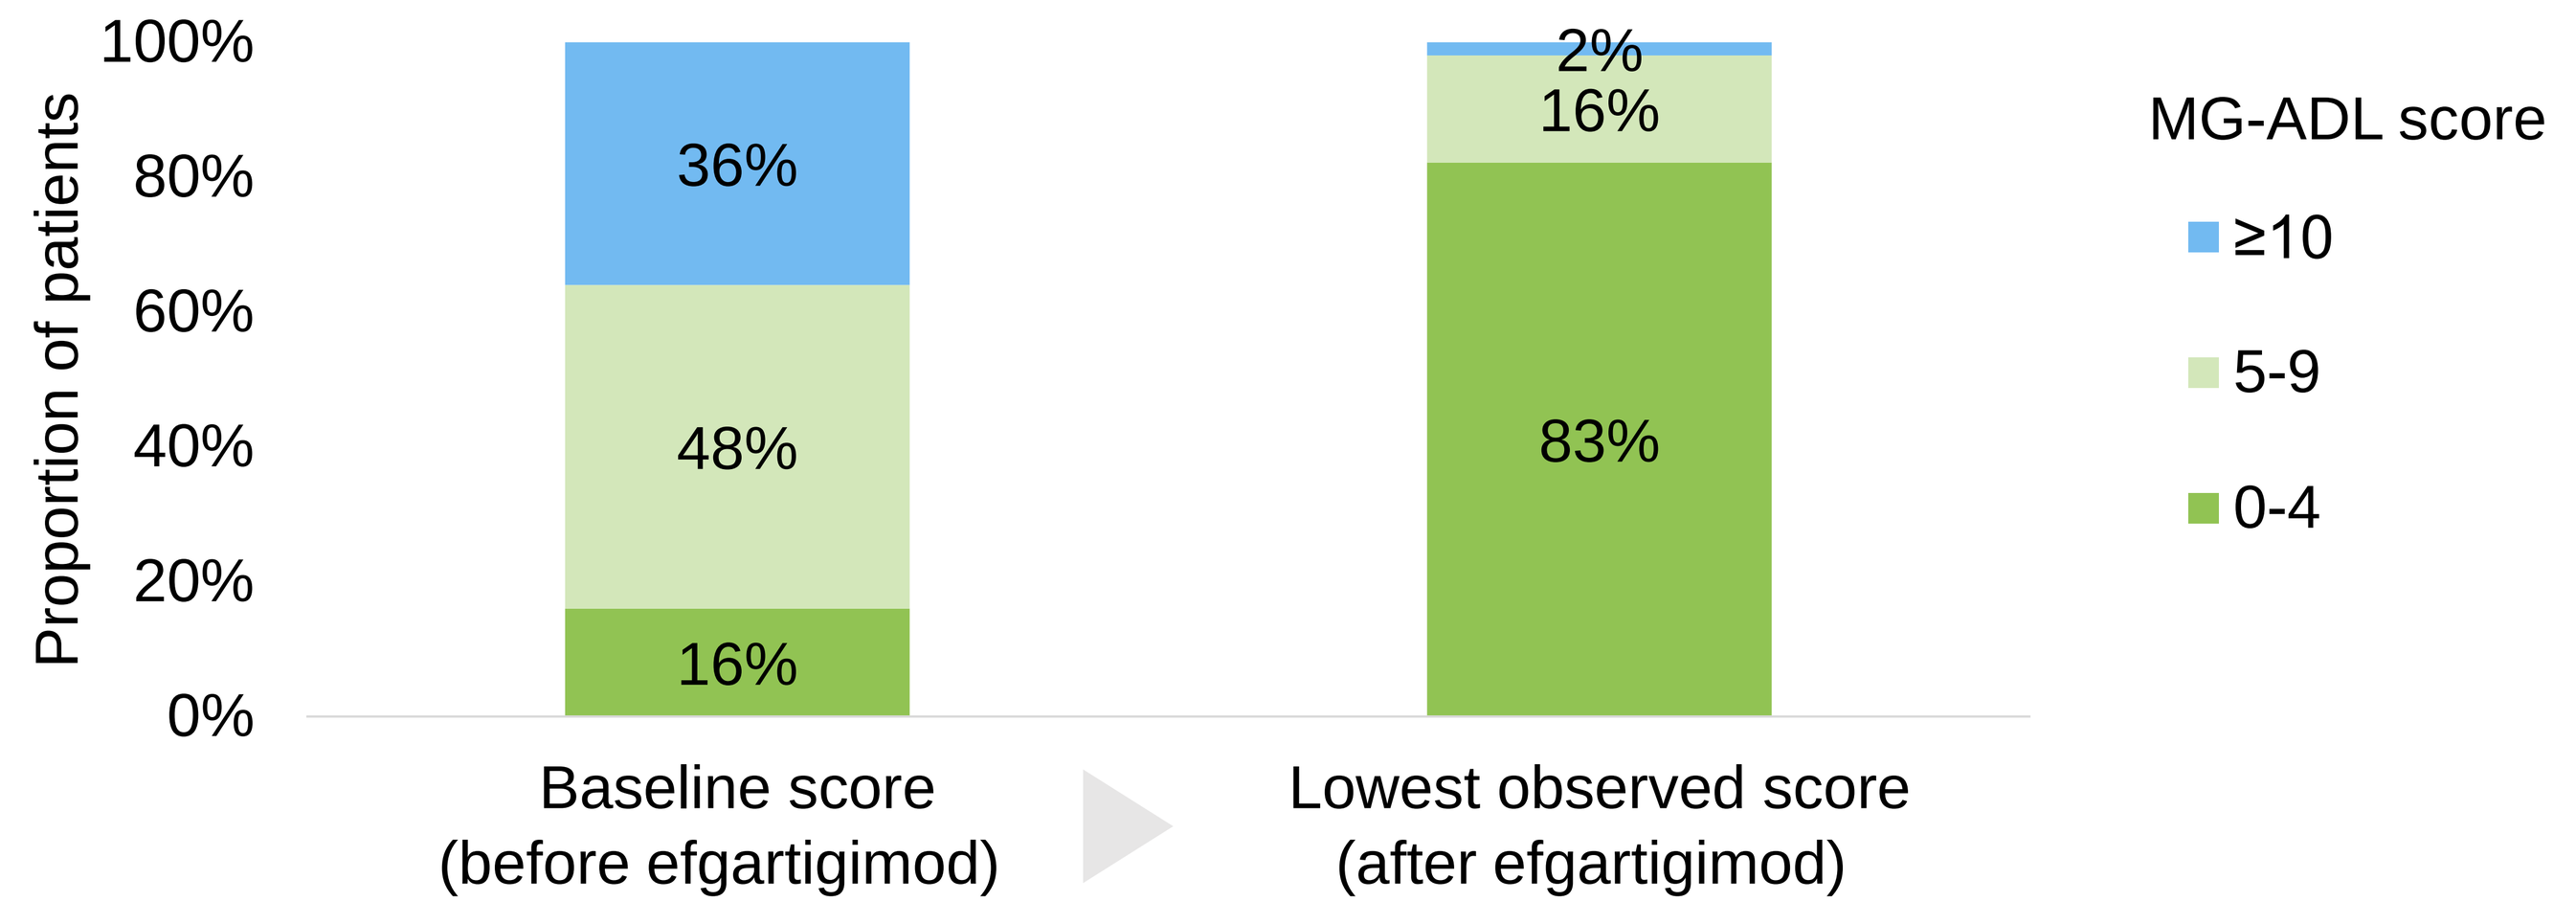

Proportion of patients
100%
80%
60%
40%
20%
0%
36%
48%
16%
2%
16%
83%
Baseline score (before efgartigimod)
Lowest observed score (after efgartigimod)
MG-ADL score
≥10
5-9
0-4

Rapid, substantial, and sustained MG-ADL benefit was observed across treatment cycles and time with efgartigimod treatment

- Among a subset of scores captured at consistent timepoints in relation to treatment cycles or time after efgartigimod initiation, MG-ADL response was rapid, substantial, and sustained, aligned with expectations based on evidence from clinical trials and clinical practice (Figures 5 and 6).

Figure 5. MG-ADL improvement by efgartigimod treatment cycle[a]

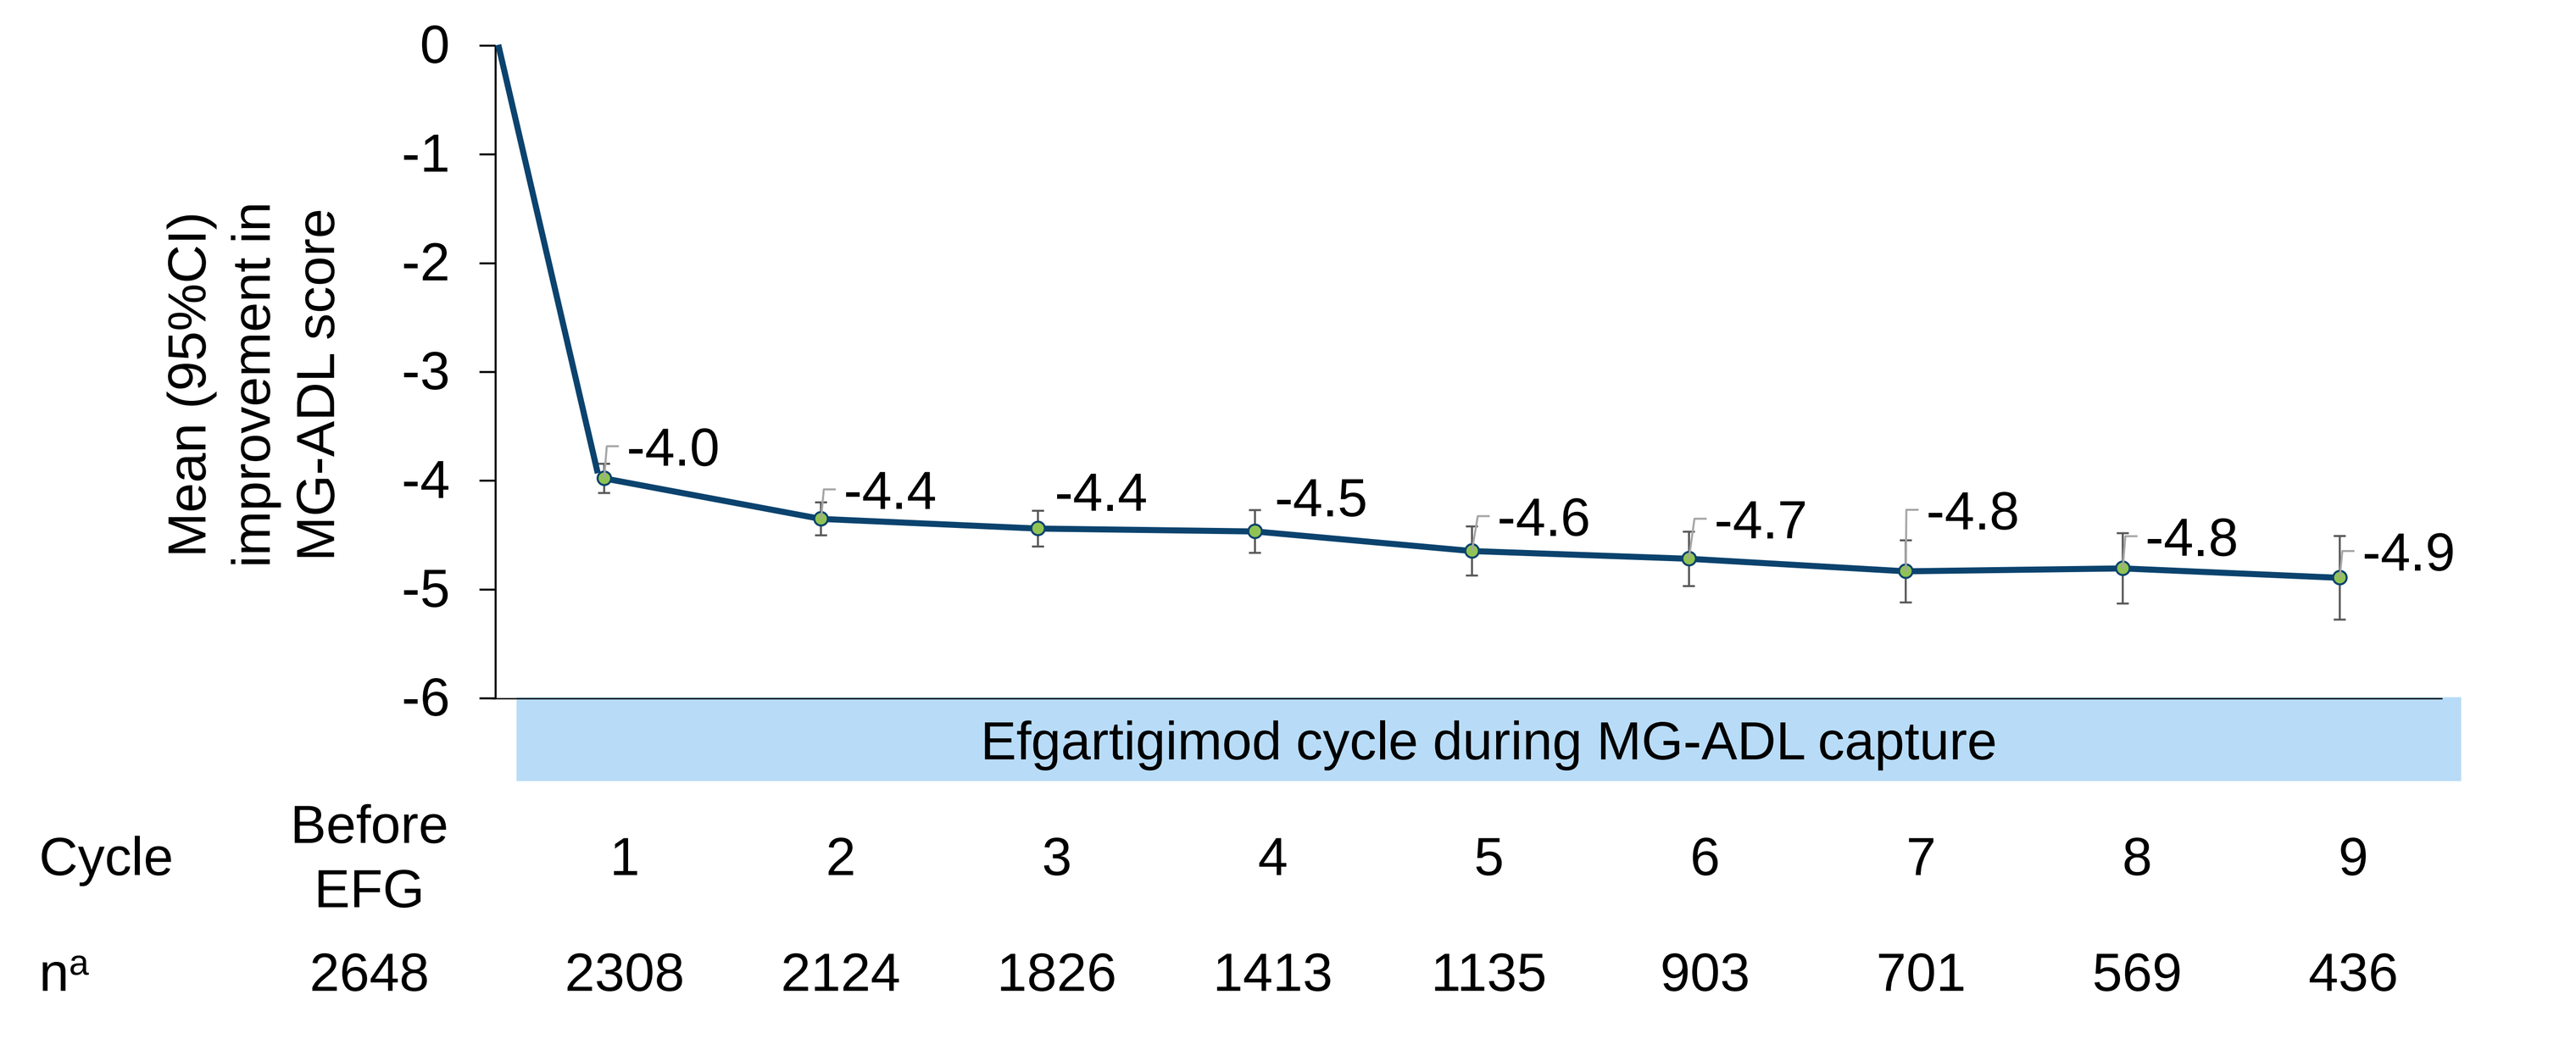

Mean (95%CI) improvement in MG-ADL score
0
-1
-2
-3
-4
-5
-6
-4.0 -4.4 -4.4 -4.5 -4.6 -4.7 -4.8 -4.8 -4.9
Efgartigimod cycle during MG-ADL capture
Cycle Before EFG 1 2 3 4 5 6 7 8 9
n[a] 2648 2308 2124 1826 1413 1135 903 701 569 436

Patients with a score recorded within any treatment cycle were included in the analysis, regardless of the cycle number. For each cycle, scores captured after the start of that cycle and before the start of the following cycle were attributed to the current cycle. For a patient's final cycle, any scores recorded after the cycle start were assigned to that cycle.

Figure 6. MG-ADL improvement by time (months) after efgartigimod initiation[a]

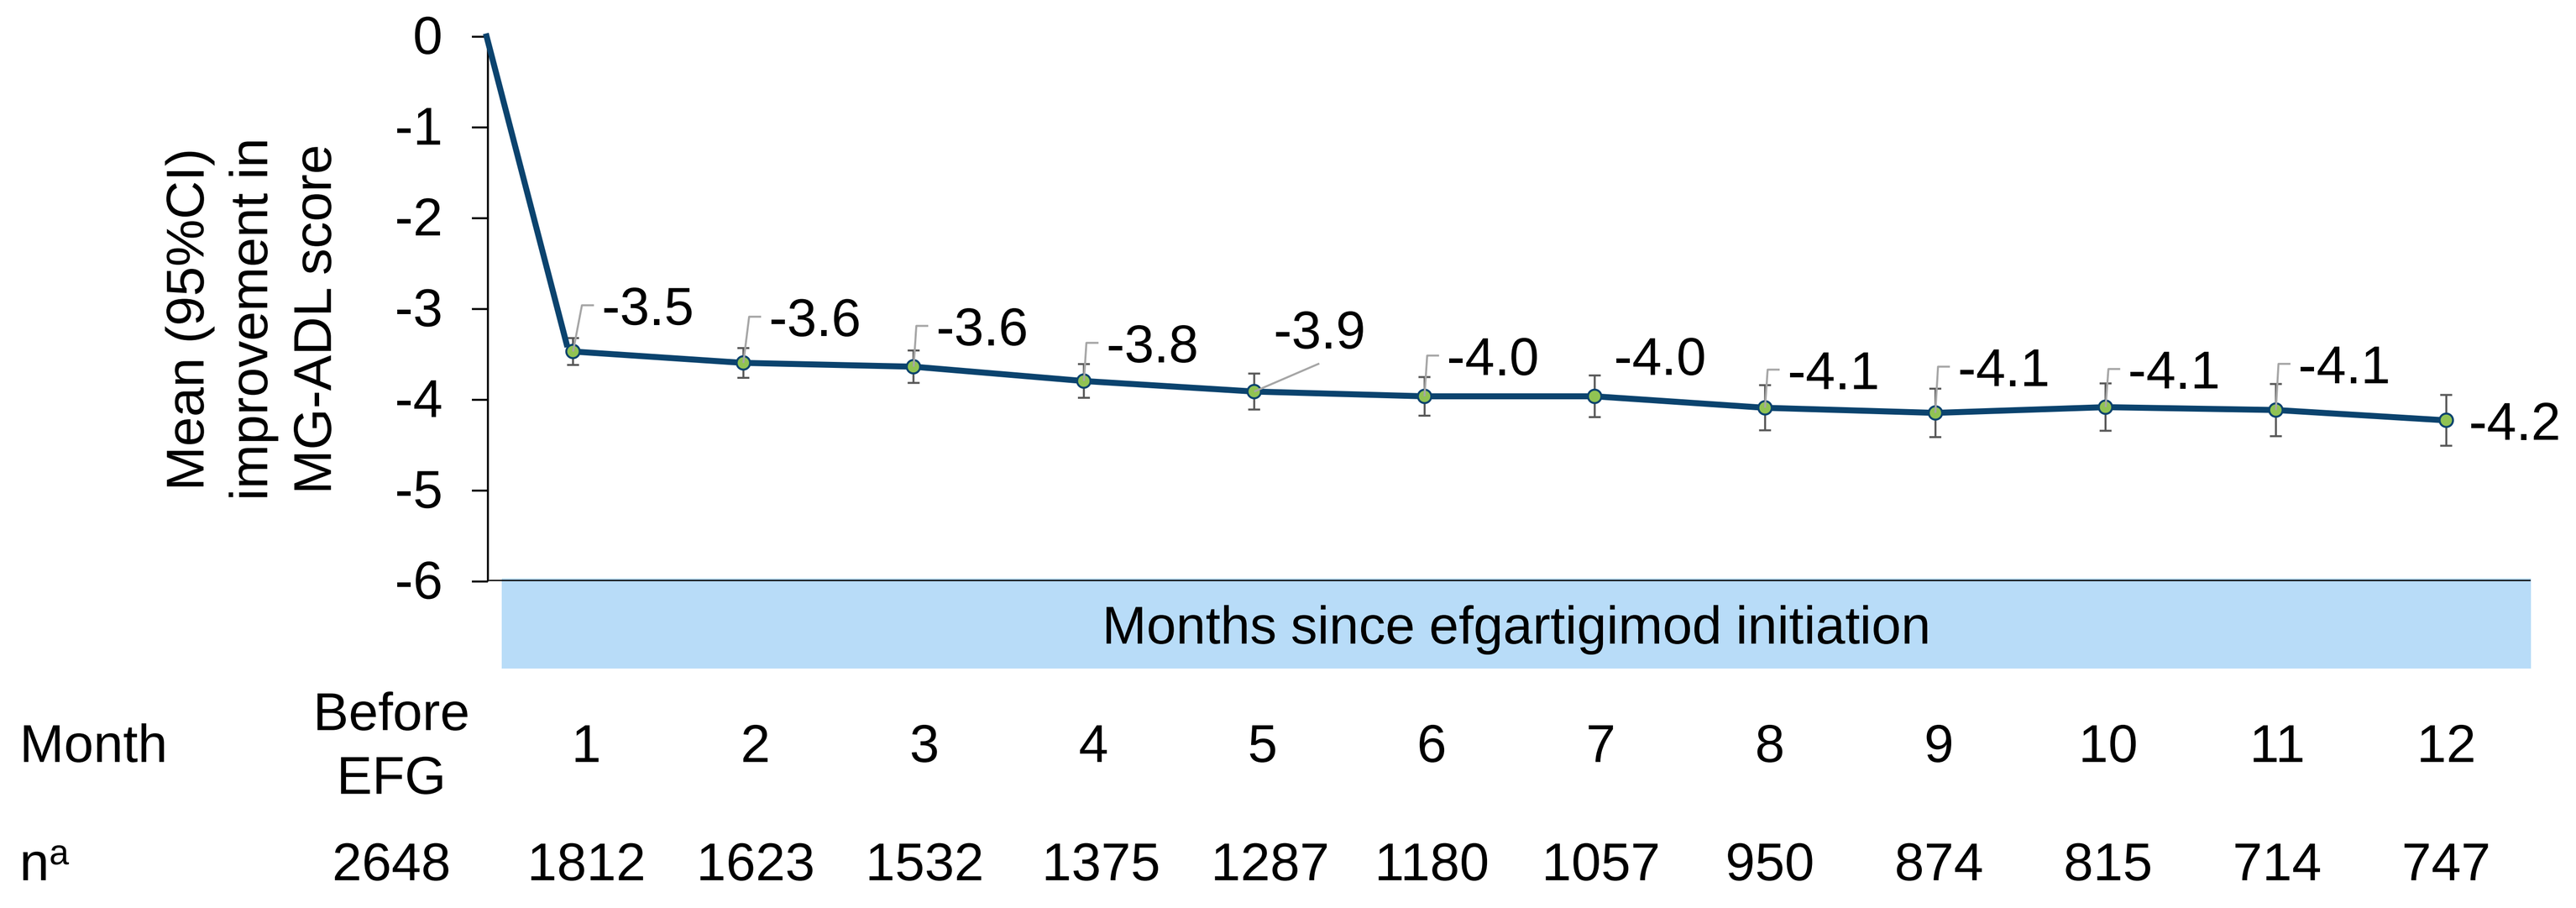

Mean (95%CI) improvement in MG-ADL score
0
-1
-2
-3
-4
-5
-6
-3.5 -3.6 -3.6 -3.8 -3.9 -4.0 -4.0 -4.1 -4.1 -4.1 -4.1 -4.2
Months since efgartigimod initiation
Month Before EFG 1 2 3 4 5 6 7 8 9 10 11 12
n[a] 2648 1812 1623 1532 1375 1287 1180 1057 950 874 815 714 747

[a]The mean change in MG-ADL was calculated as the difference between the baseline score and the lowest (best) MG-ADL score recorded during the specific month after efgartigimod initiation. At each month, only patients who had any MG-ADL score captured were included.

ACKNOWLEDGMENTS AND DISCLOSURES: This study was funded by argenx US, Inc. **PN** has received research support from APCORI, Alexion/AstraZeneca Rare Disease, Momenta/Janssen, and Ra/UCB, Dianthus, and served as member of advisory boards or provided paid consultations to Alexion/Astra Zeneca Rare disease, Amgen, Argenx, CVS, Dianthus, GSK, ImmuneAbs, Janssen, Novartis, and UCB, and has been the Data Monitoring Committee Chair at Sanofi and argenx, and received royalties from Springer Nature. **MO, CQ, MJ, EB, and DG** are employees of argenx. **RB-S** has served on advisory boards for argenx and is a consultant to Sanofi. **AGS** has served as a member of advisory boards or paid consultant for Alexion, Arcellx, argenx, Eidos, Lexicon, Lilly, Seismic, and UCB. **RRM and MS** are employees of ZS Associates and serve as paid consultants for argenx. **GIW** has served as member of advisory boards or provided paid consultations to Alexion, argenx, Cartesian, Janssen, and UCB, is on speaker bureaus for Alexion and UCB, and has received research support from Alexion, argenx, Immunovant, Roche, UCB, and the MG Foundation of America.

REFERENCES: 1. argenx. VYVGART (efgartigimod alfa-fcab) Prescribing Information. United States Food and Drug Administration. Updated Aug 2024. Accessed Mar 28, 2025. **2.** Howard JF Jr, et al. *Lancet Neurol*. 2021;20(7):526-536. **3.** European Medicines Agency. Vyvgart efgartigimod alfa (2022). Accessed 18 Jul 2024. **4.** Suzuki S, et al. *Neurol Clin Pract*. Jul 2024;14(3):e200276. **5.** argenx. VYVGART Product Monograph. Health Canada Drug Product Database. Updated Sep 19, 2023. Accessed Mar 7, 2025. **6.** Zhu G, et al. *Ther Adv Neurol Disord*. 2025;18:17562864241311127. **7.** Silvestri NJ. *Muscle Nerve*. Mar 2025;71(3):422-428. **8.** Singer M, et al. *Muscle Nerve*. Nov 21 2023. **9.** Frangiamore R, et al. *Eur J Neurol*. Apr 2024;31(4):e16189. **10.** Jin L, et al. *Ther Adv Neurol Disord*. 2025;18:17562864251319656. **11.** Fuchs L, et al. *J Neurol*. Mar 25 2024.

argenx

15th Myasthenia Gravis Foundation of America (MGFA) International Conference on Myasthenia and Related Disorders | May 13–15, 2025 | The Hague, The Netherlands